Immuno-stimulatory activities of blends of natural extracts in human immune cells

Hong Wang, M.D., Ph.D. and Bruce Daggy, Ph.D.
R &D, Shaklee Corporation, Pleasanton, CA 94588

Conflict of Interest:

Authors are Shaklee Corporation employees.

ABSTRACT

Polysaccharides are naturally present as part of the cell wall components in plant and fungi. Certain polysaccharides with specific structures such as β-D- (1, 3/1, 6) -glucan, are the active ingredients in many well-known immuno-modulatory foods including yeast and mushrooms. β-D- (1,3/1,6)-glucan is recognized and are taken up by the mammalian immune cells such as macrophages/dendritic cells via beta-glucan receptors (dectin-1/TLR-2) on their cell membrane. Due to these polysaccharides are similarly found in microorganisms, our immune cells regard them as "pathogen-associated molecules" and will elicit an activated immune response to prepare our body for defense. Previously we investigated the in vitro immuno-stimulatory effects on cytokine production and lymphocyte proliferation of a blend of extracts containing polysaccharides including yeast beta-glucan in human immune cells isolated from blood of a healthy donor. The results showed that the blend had potent immuno-stimulatory effects and was superior to β-glucan alone (10 – 100 µg/mL) in several cytokine parameters (IFN-γ, IL-10, IL-12) and lymphocyte proliferation index. We further explored other potential blends of natural extracts with that were known to have immune enhancing properties. Interestingly, while some combination showed synergistic enhancement in activities, others showed reduced efficacy compared to β-glucan (10 µg/mL) alone. Based on our results, we were able to develop a dietary supplement that has the optimal combination of natural extracts to potentiate the immuno-stimulatory effects of β-glucan and enhance the efficacy.

METHODS

Lymphocyte proliferation

Blood was obtained from a single healthy donor and peripheral blood mononuclear cells (PBMCs) were prepared by centrifugation over a ficoll hypaque density gradient, followed by washing. Viable PBMCs were adjusted to a concentration of 2 x 10^6/mL in complete medium consisting 100 µL/well were placed in triplicate in 96-well microtiter plates. Following this, 100 µL of complete medium, complete medium containing 1% DMSO (background controls), various concentrations of the compounds were added. The final DMSO concentration was kept constant at 0.5% for the DMSO-soluble compounds and the accompanying background control wells.

Cultures was incubated at 37ºC in 5% CO_2 for 3 days, pulsed with 1 µCi of tritiated thymidine (H3-TdR) for the final 6-16 hours, harvested, and counted to determine H3-TdR incorporation (a readout of proliferation).

Cytokine Production

PBMCs were prepared as described above and 100 µL/well was placed in triplicate in 96- well microtiter plates. Following this, 100 µL of complete medium, complete medium containing 1% DMSO (background controls), various concentrations of the compounds were added. After approximately 24 hours of incubation at 37ºC in 5% CO_2, culture supernatants (SNs) were removed and frozen. The SNs were analyzed by multiplexing in the Luminex 200 System for various cytokines.

RESULTS

Figure 1.

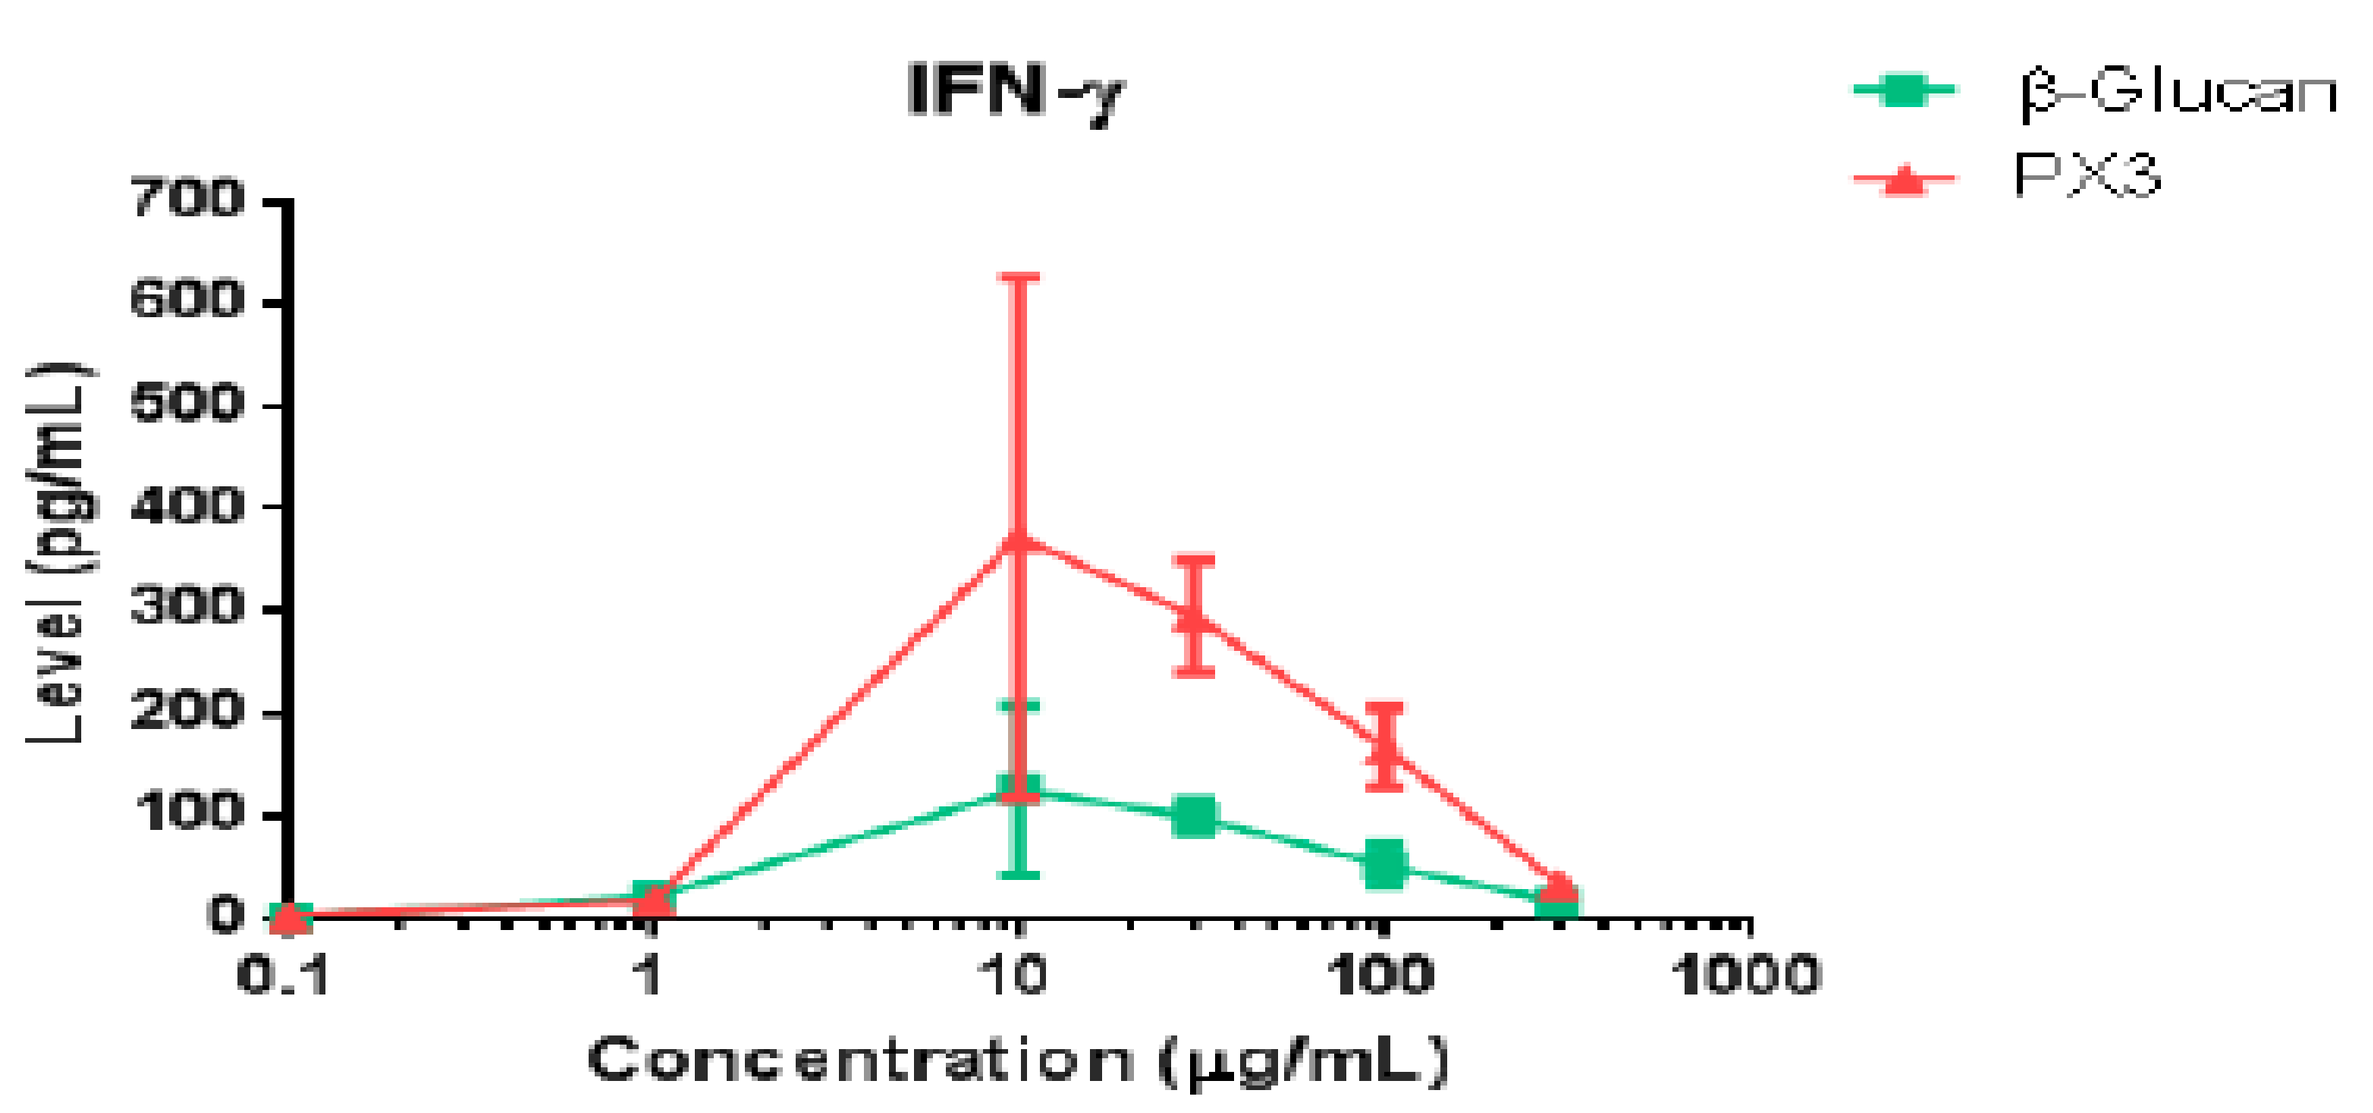
IFN-γ
β-Glucan
PX3
Level (pg/mL)
Concentration (µg/mL)

Two way ANOVA:
** Group p= 0.0047
**** Concentration p <0.0001
* Interaction p< 0.0488

Figure 2.

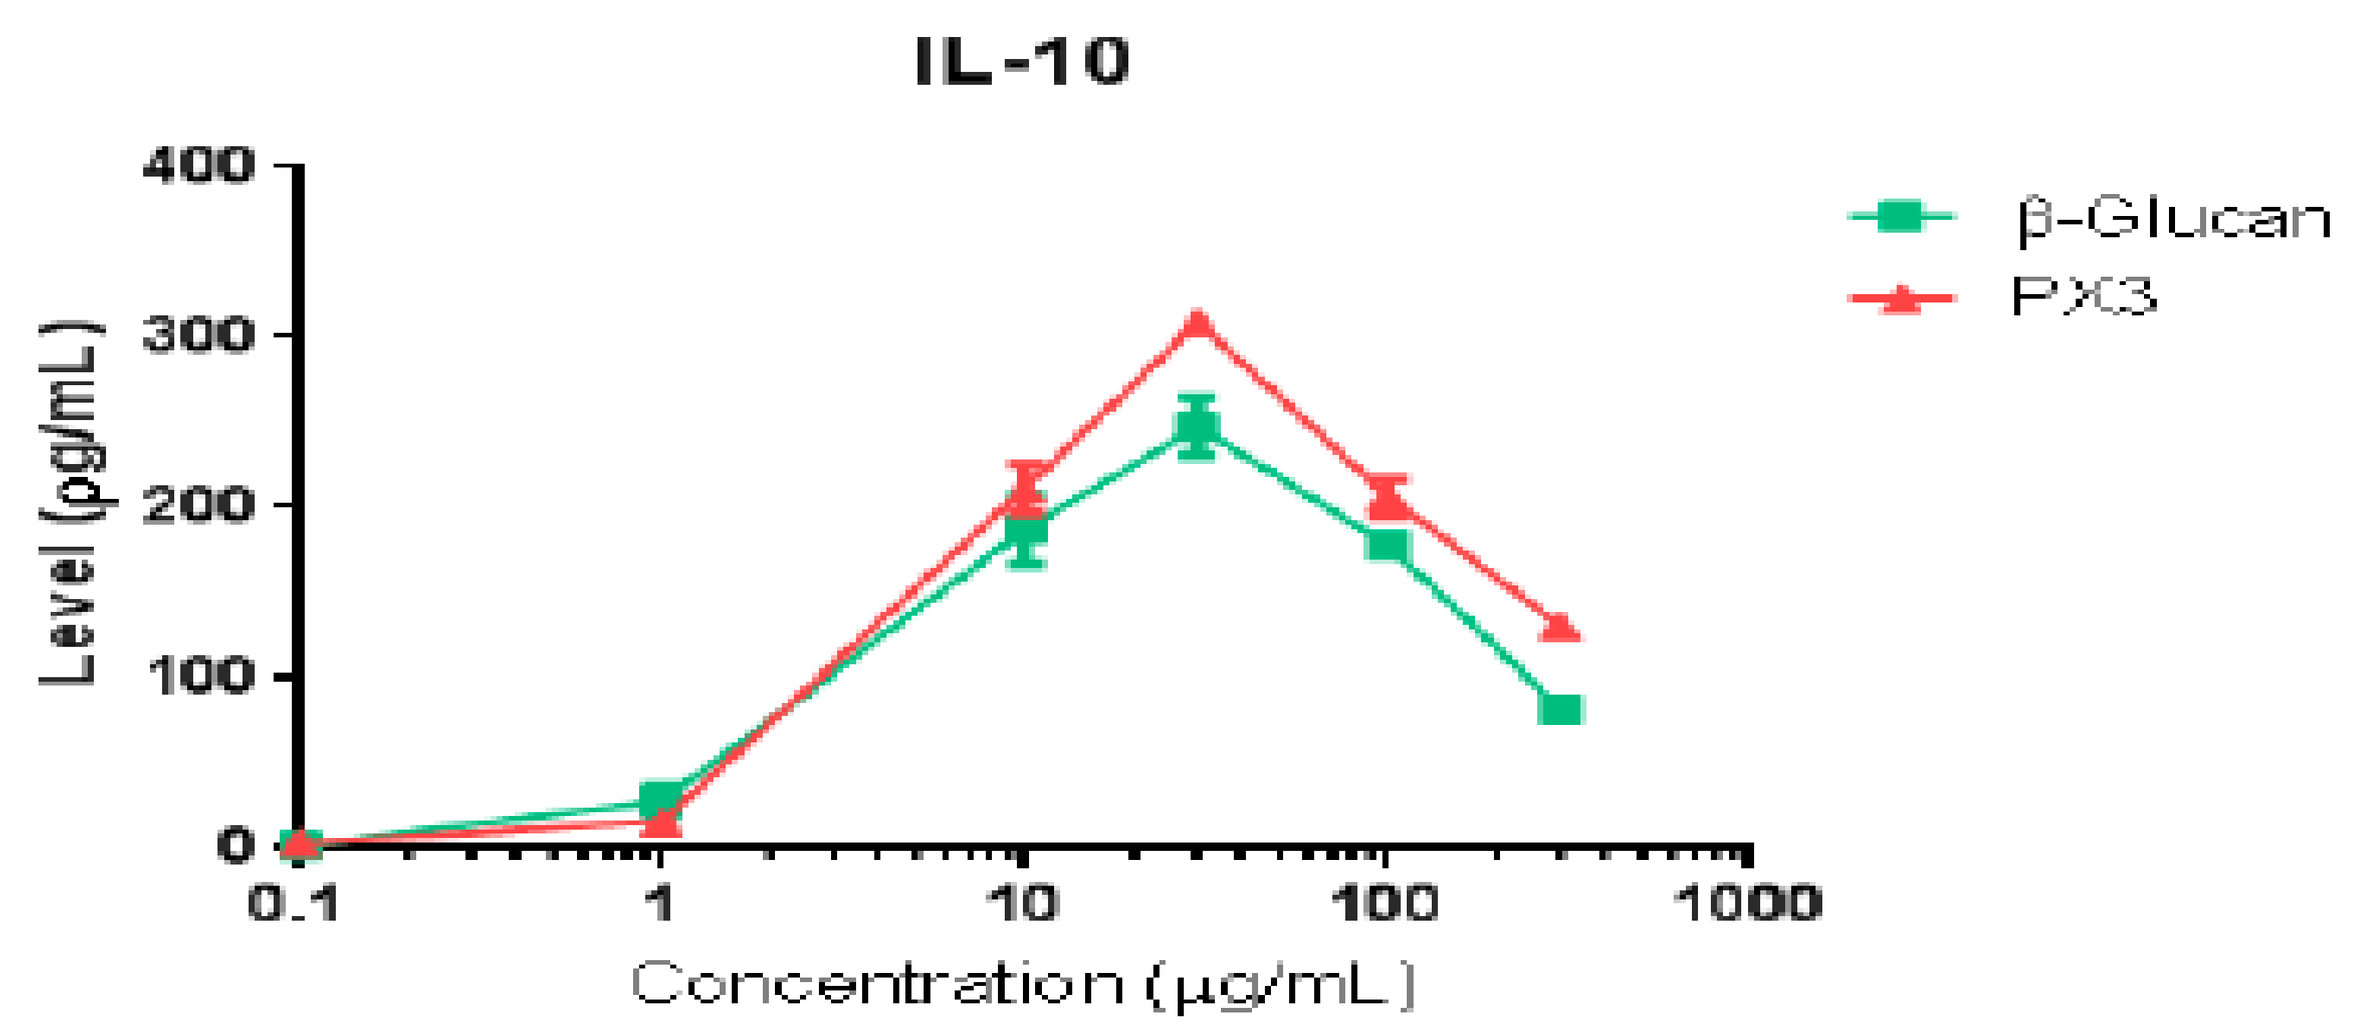
IL-10
β-Glucan
PX3
Level (pg/mL)
Concentration (µg/mL)

Two way ANOVA:
**** Group p <0.0001
**** Concentration p <0.0001
**** Interaction p <0.0001

Figure 3.

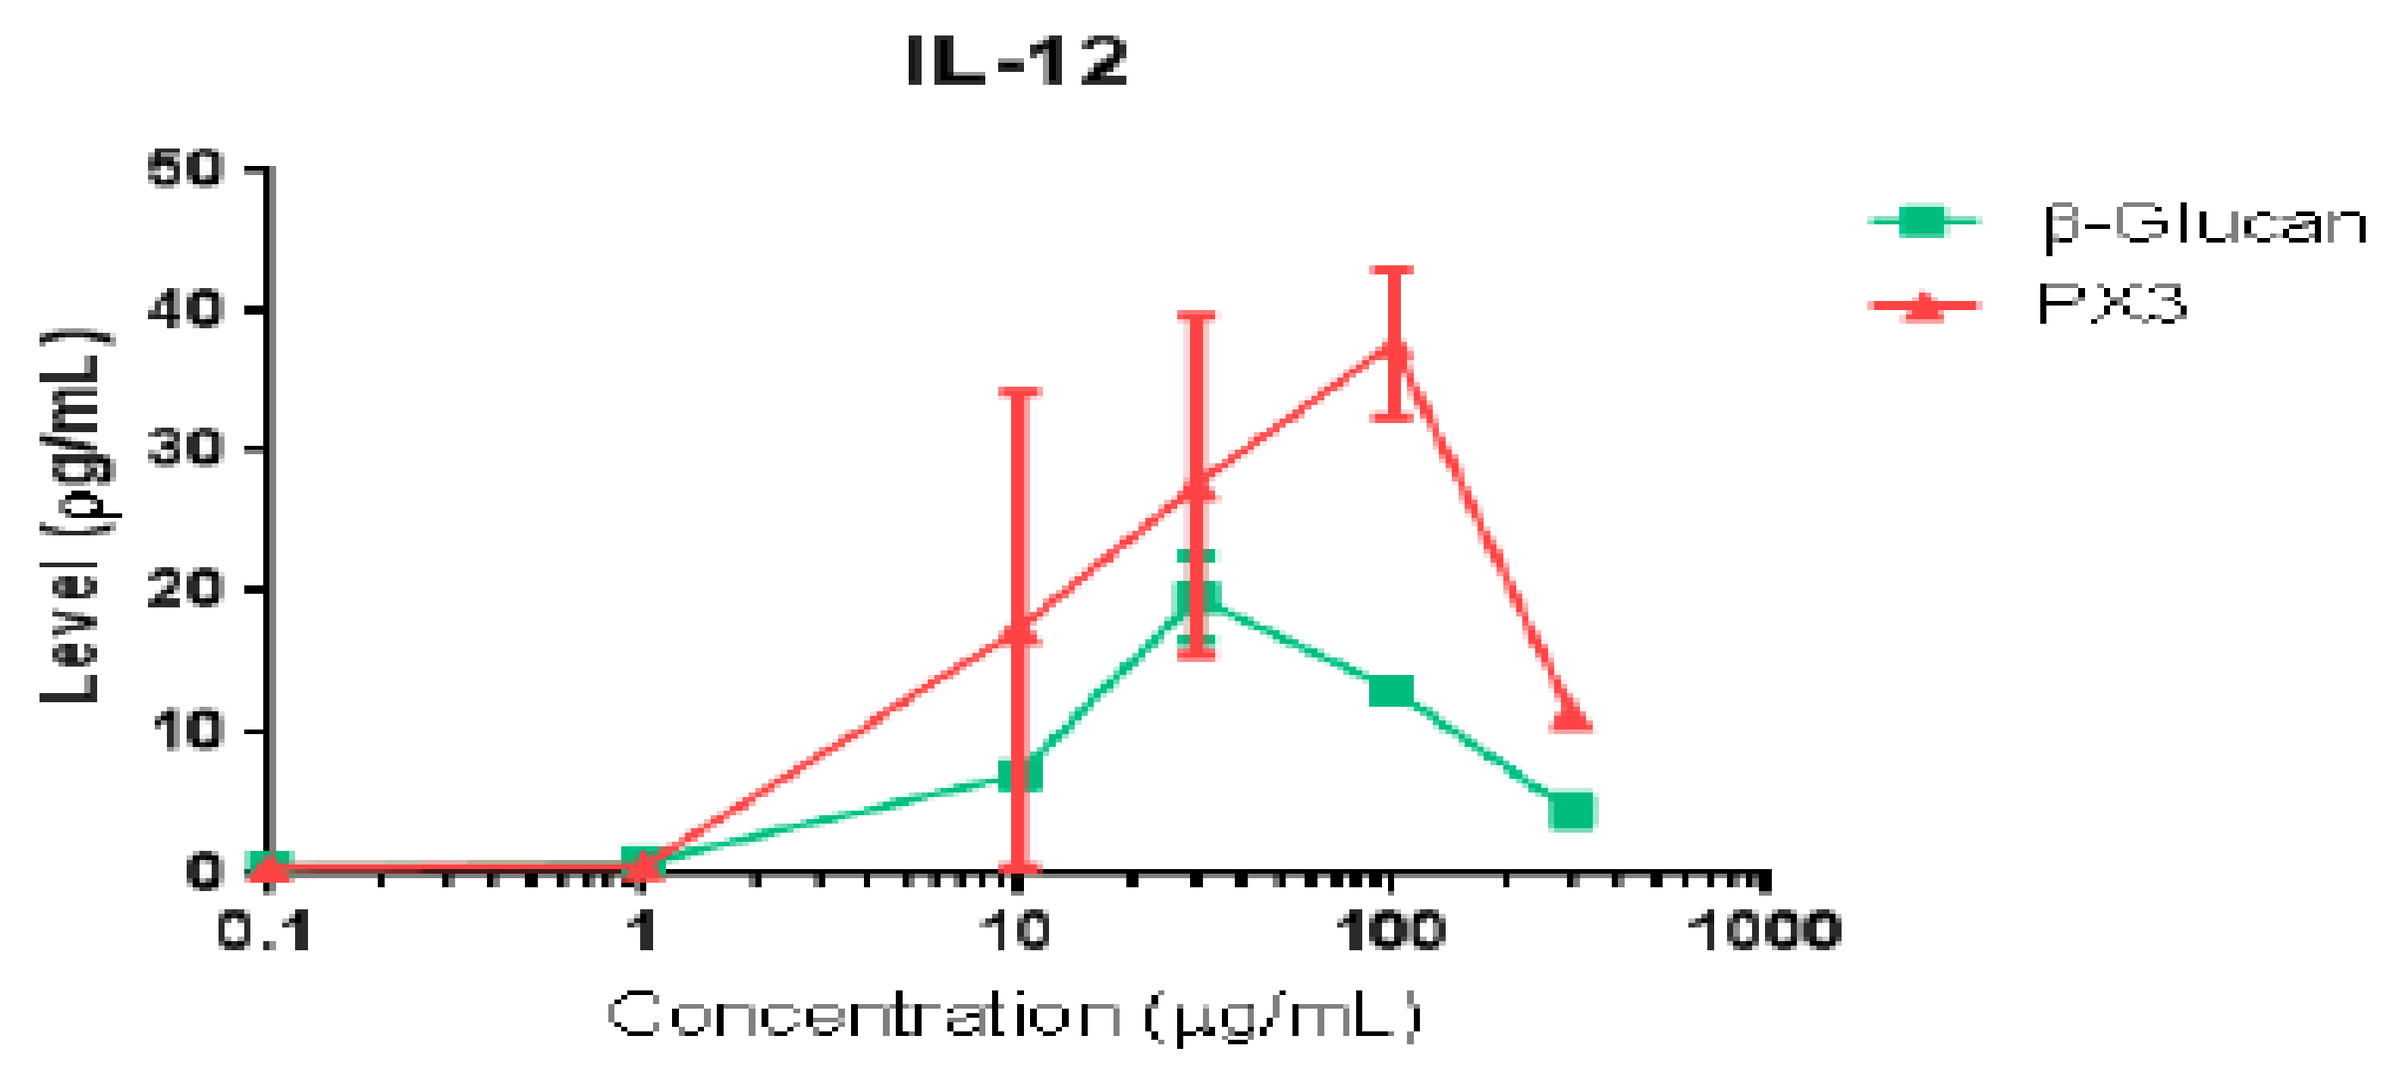
IL-12
β-Glucan
PX3
Level (pg/mL)
Concentration (µg/mL)

Two way ANOVA:
** Group p = 0.002
**** Concentration p <0.0001
* Interaction p = 0.0264

Figure 4.

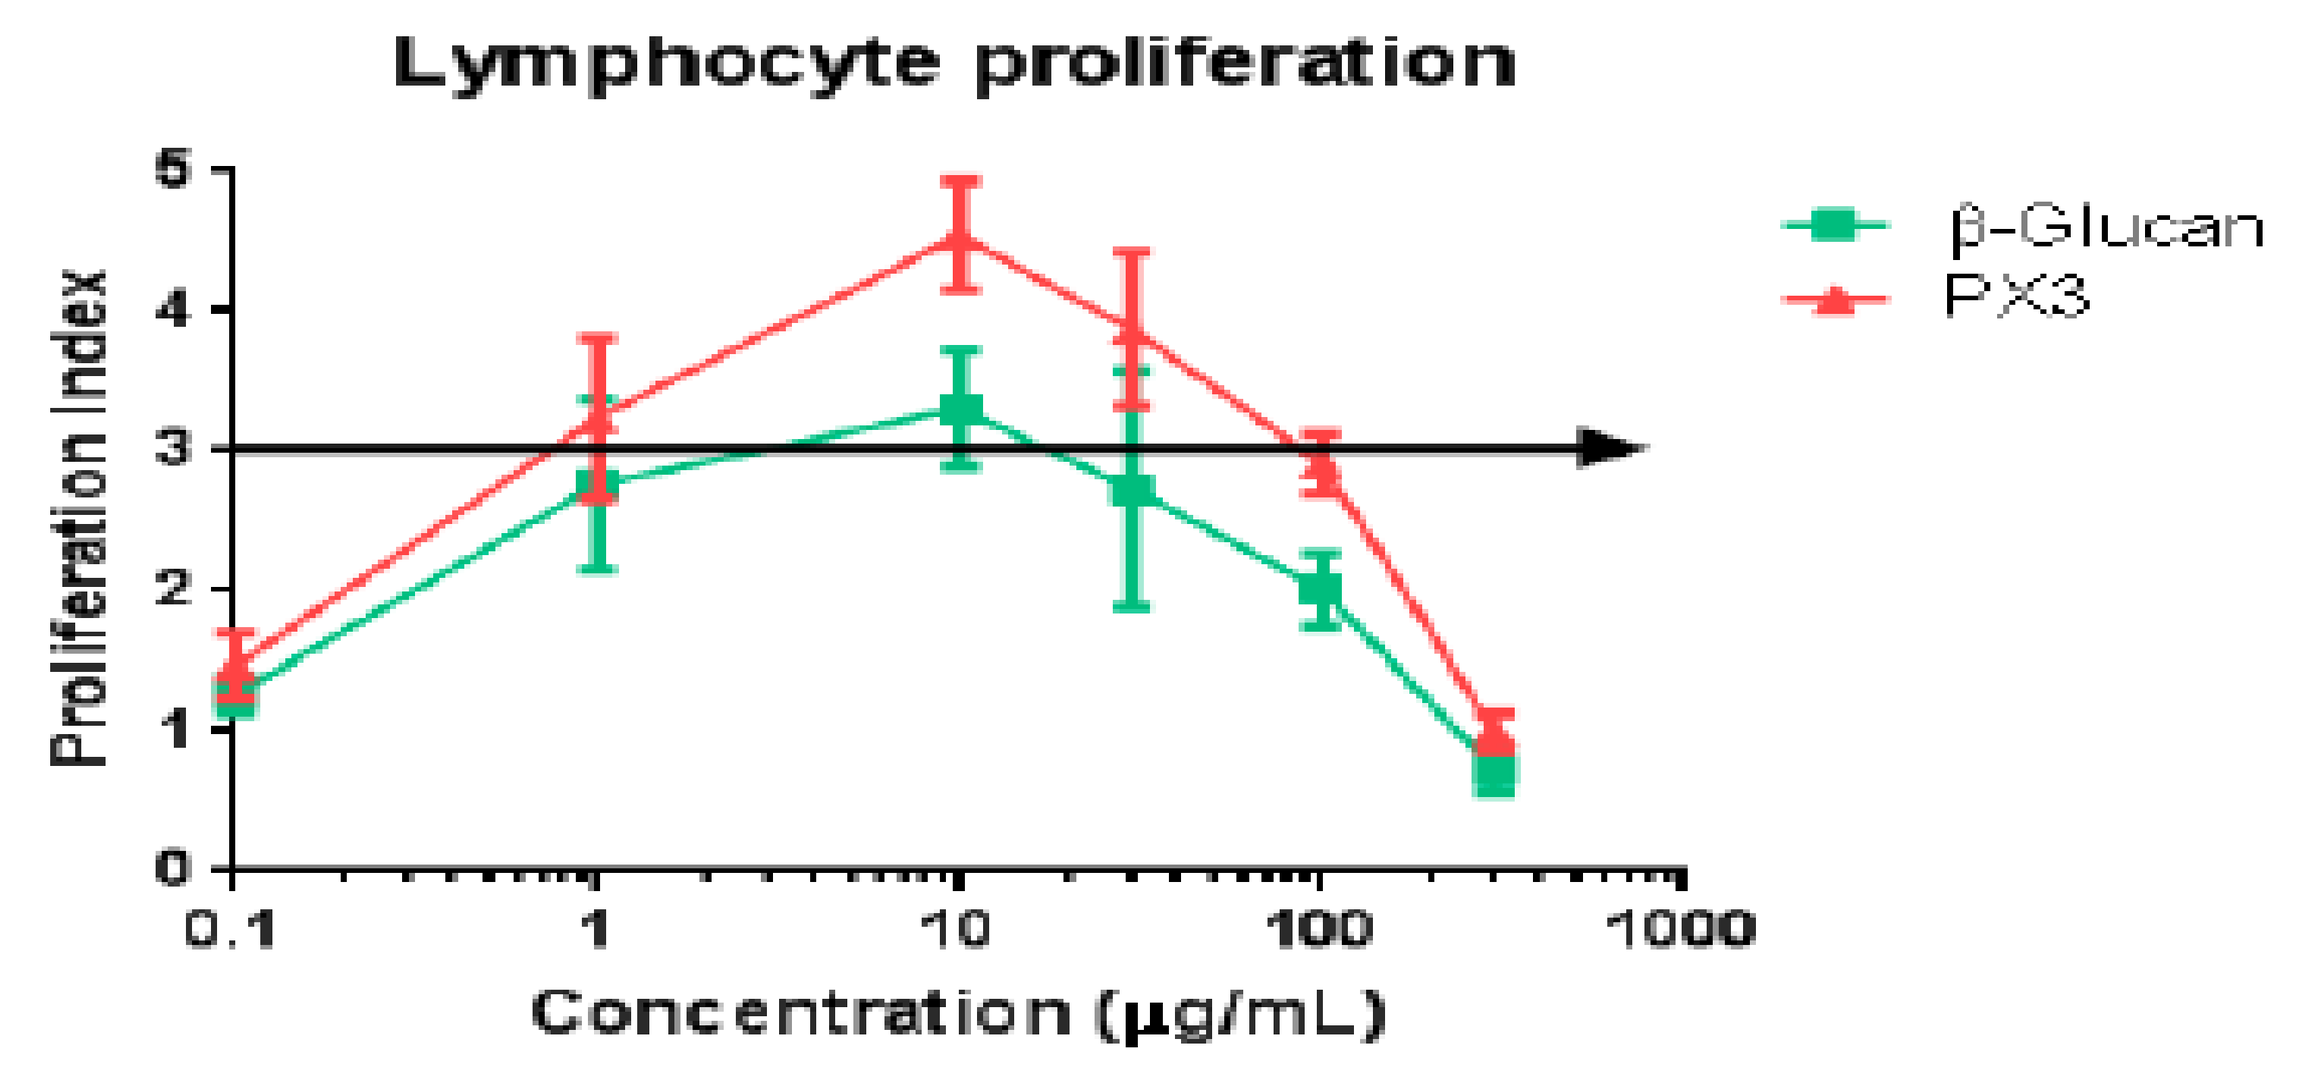
Lymphocyte proliferation
β-Glucan
PX3
Proliferation Index
Concentration (µg/mL)

Two way ANOVA:
**** Group p <0.0001
**** Concentration p <0.0001
Interaction p= 0.077

Immuno-stimulatory activities of blends of natural extracts in human immune cells

Hong Wang, M.D., Ph.D. and Bruce Daggy, Ph.D.

R &D, Shaklee Corporation, Pleasanton, CA 94588

Conflict of Interest:

Authors are Shaklee Corporation employees.

RESULTS CONTINUED

Figure 5.

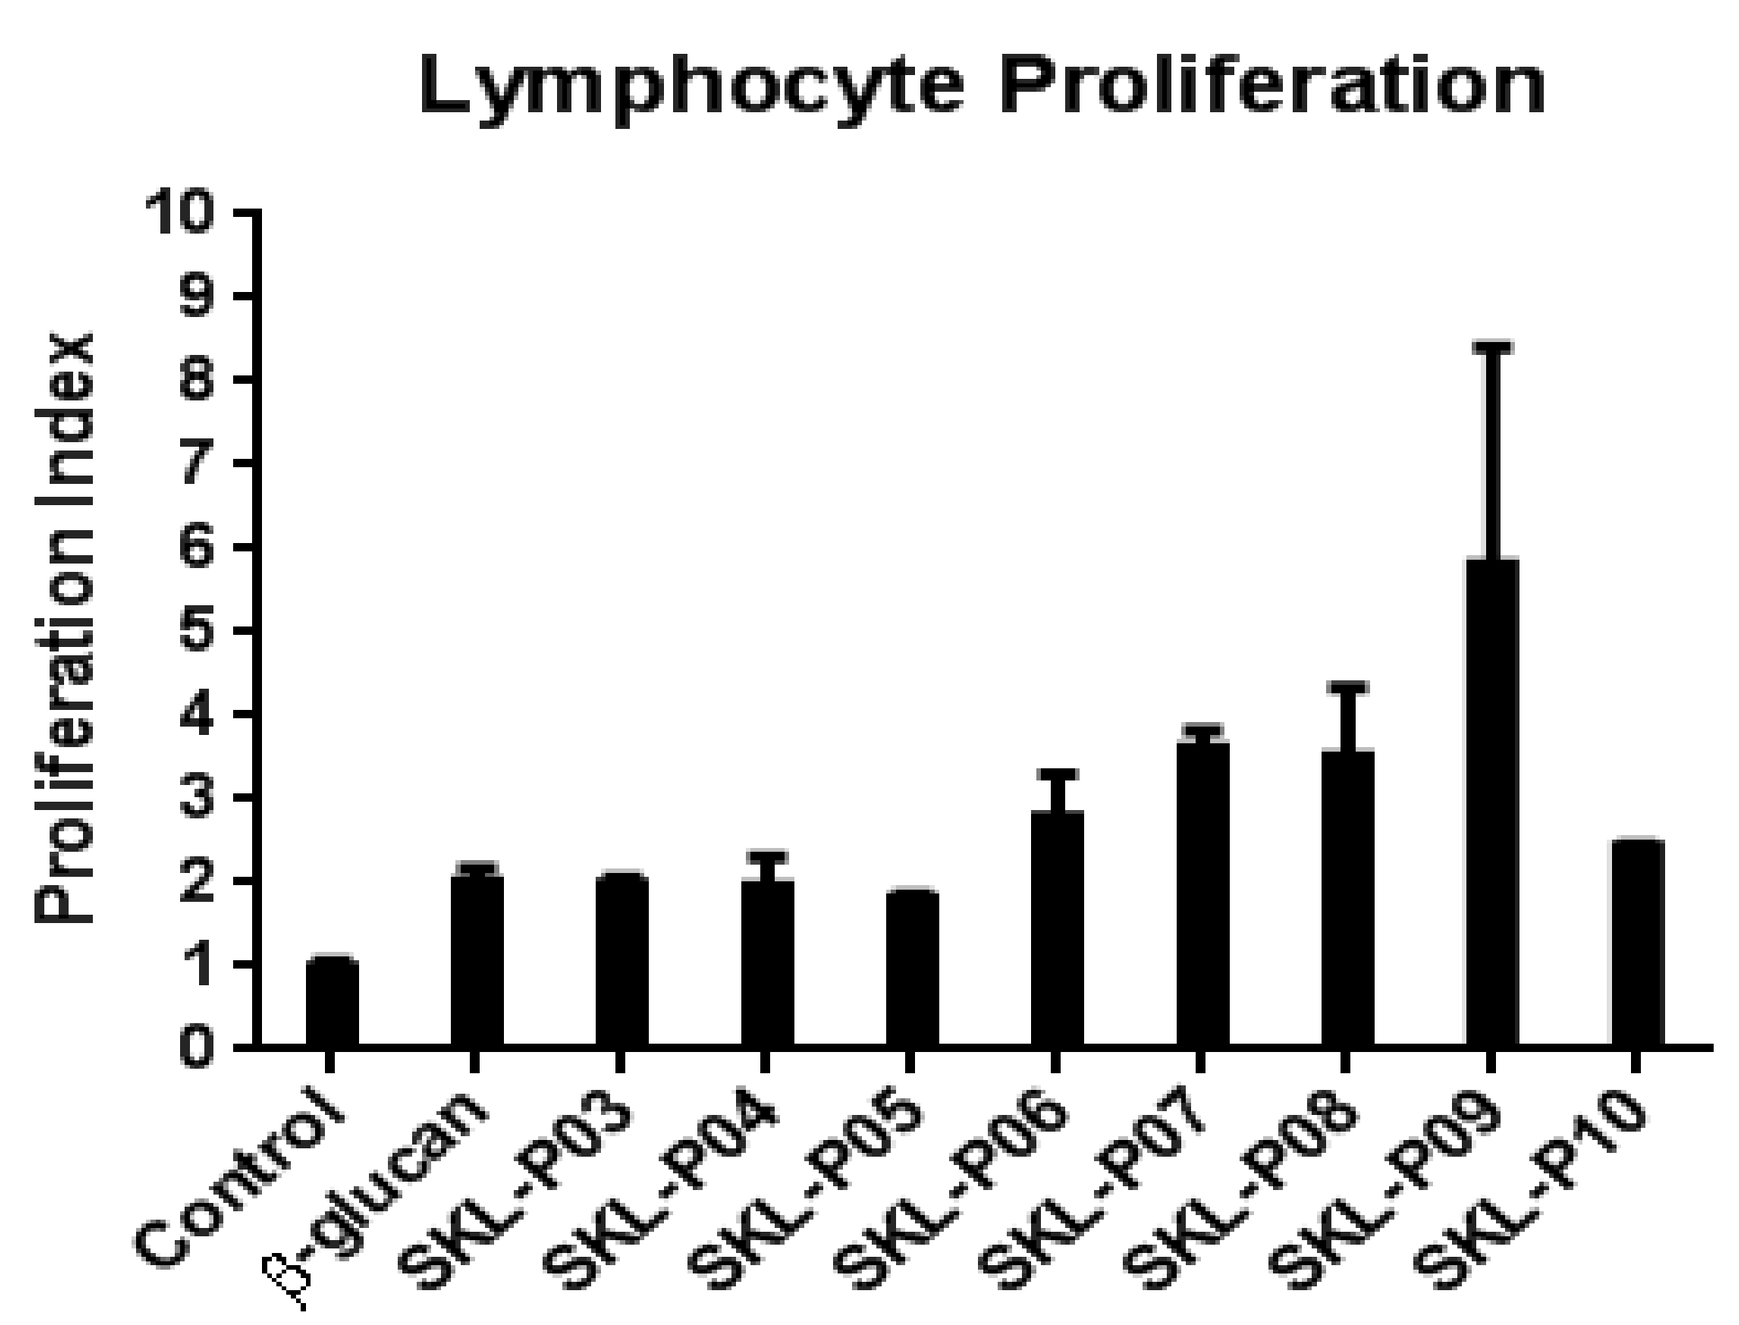
Lymphocyte Proliferation
Proliferation Index
0 1 2 3 4 5 6 7 8 9 10
Control
β-glucan
SKL-P03
SKL-P04
SKL-P05
SKL-P06
SKL-P07
SKL-P08
SKL-P09
SKL-P10

Figure 6.

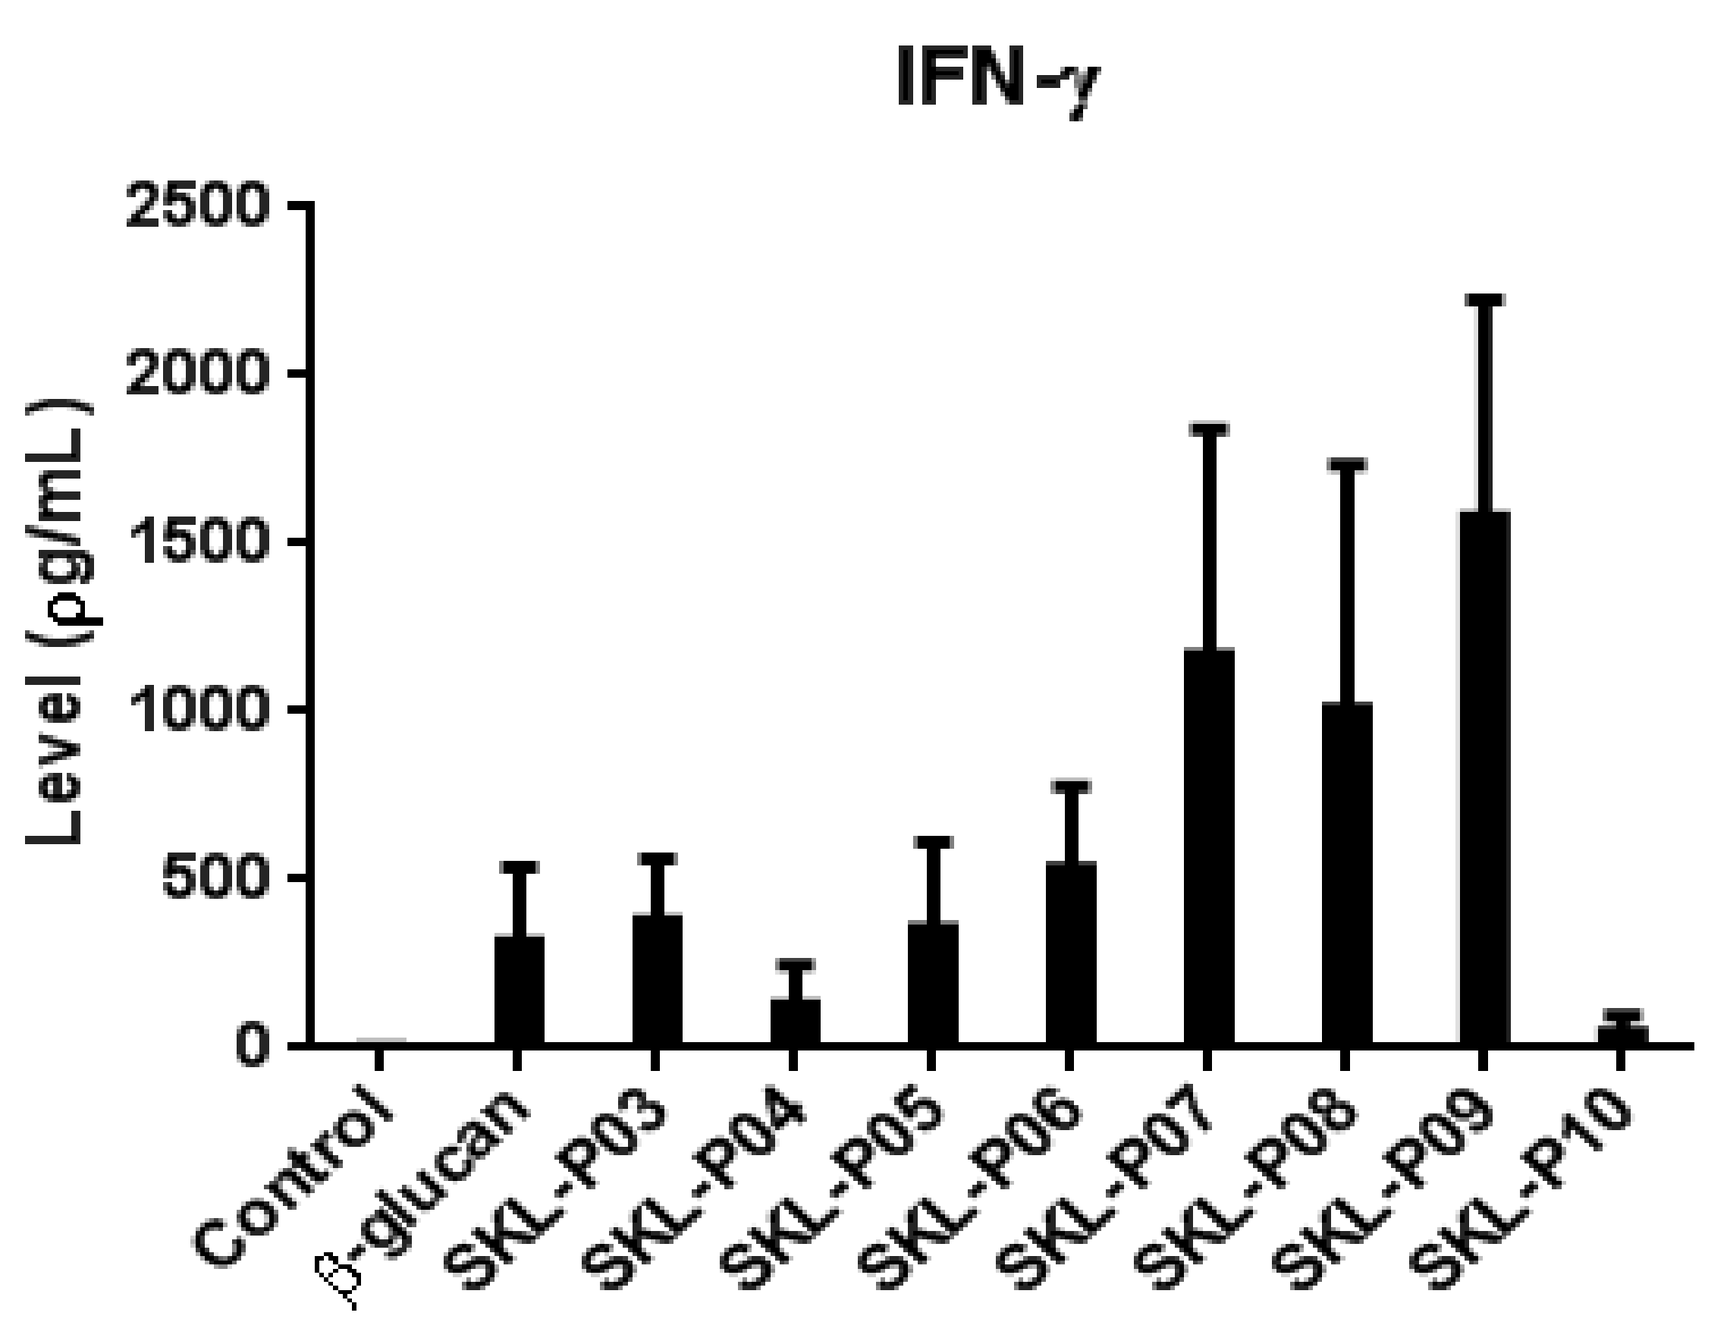
IFN-γ
Level (ρg/mL)
0 500 1000 1500 2000 2500
Control
β-glucan
SKL-P03
SKL-P04
SKL-P05
SKL-P06
SKL-P07
SKL-P08
SKL-P09
SKL-P10

Figure 7.

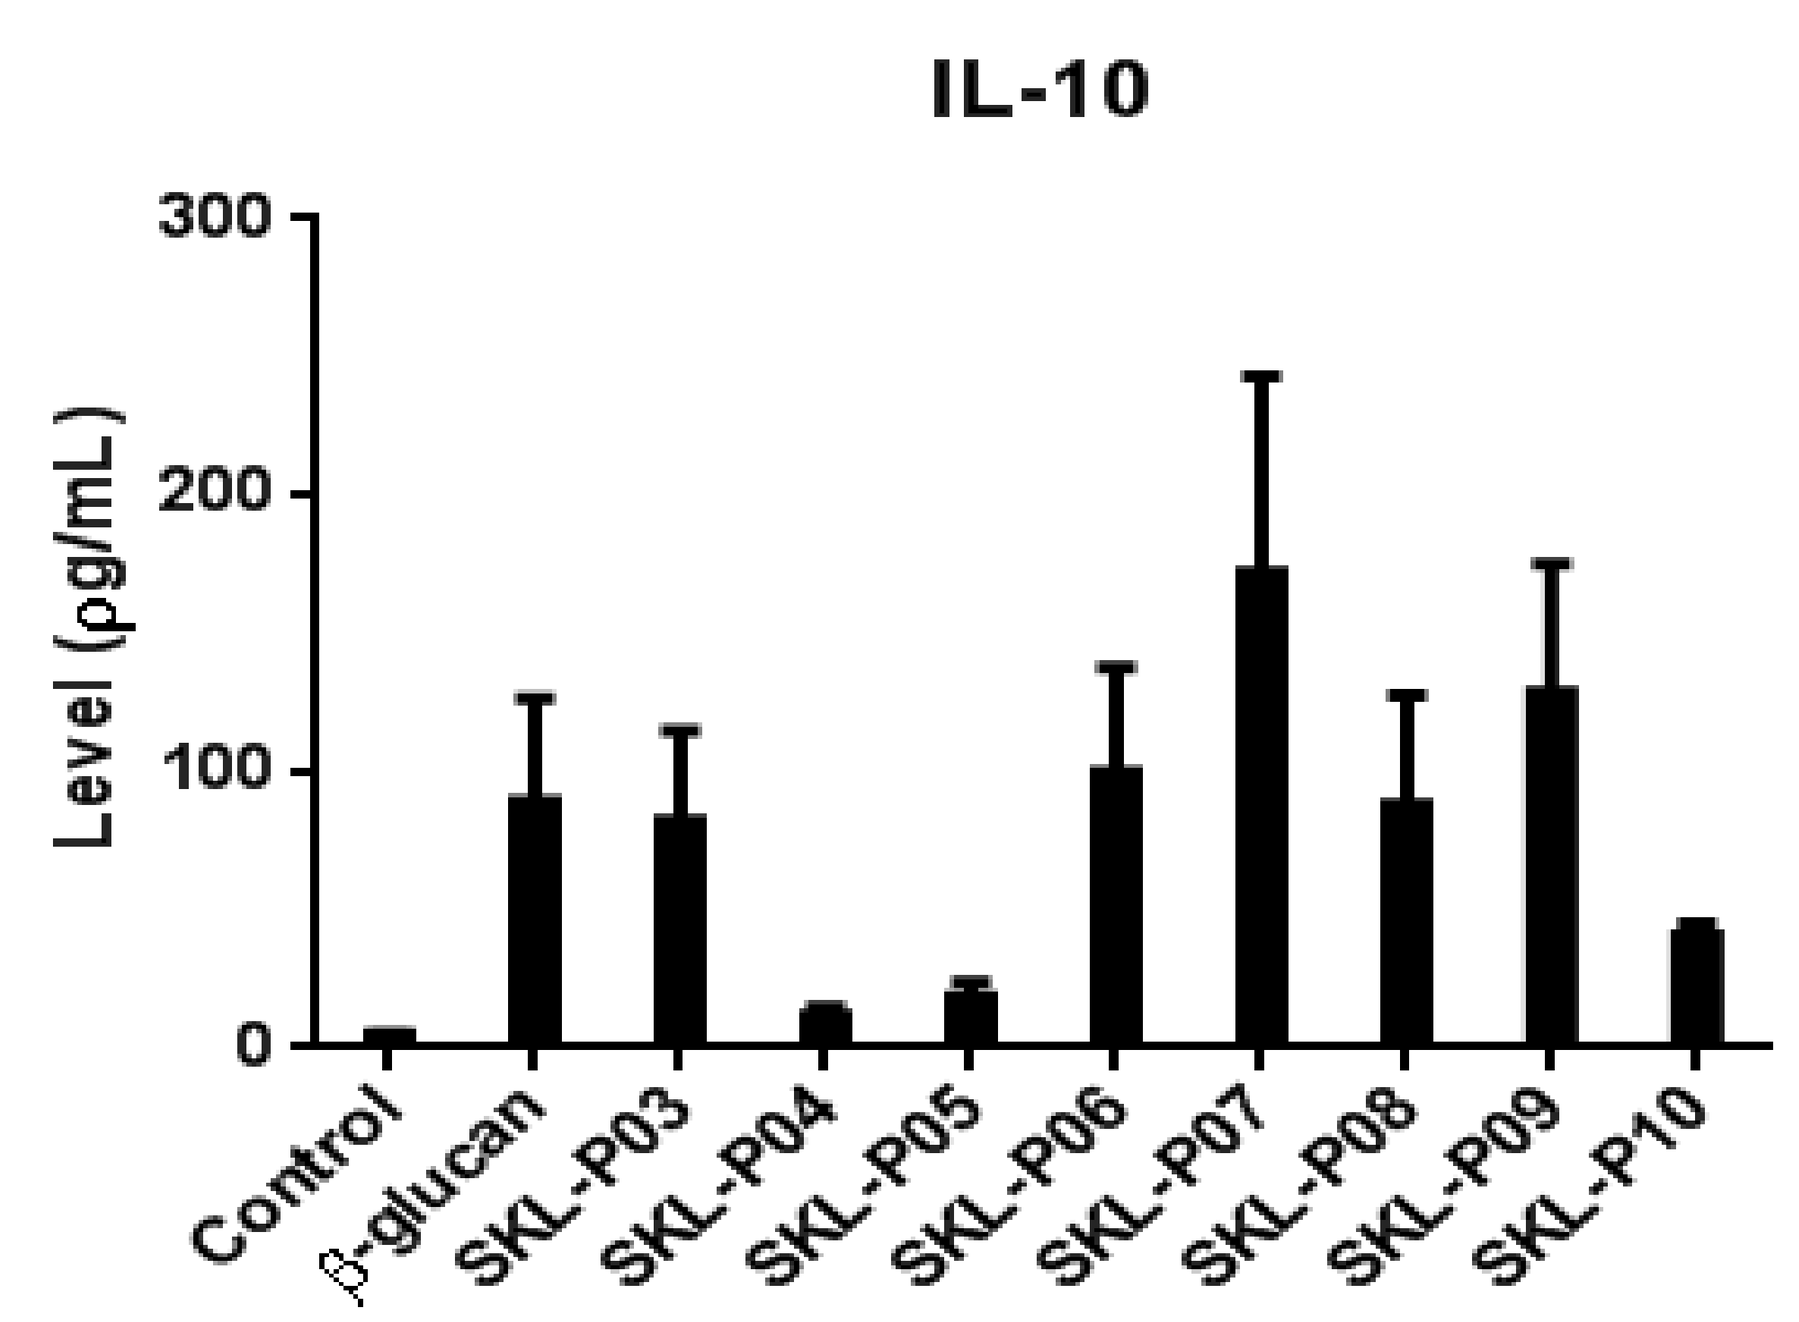
IL-10
Level (ρg/mL)
0 100 200 300
Control
β-glucan
SKL-P03
SKL-P04
SKL-P05
SKL-P06
SKL-P07
SKL-P08
SKL-P09
SKL-P10

Figure 8.

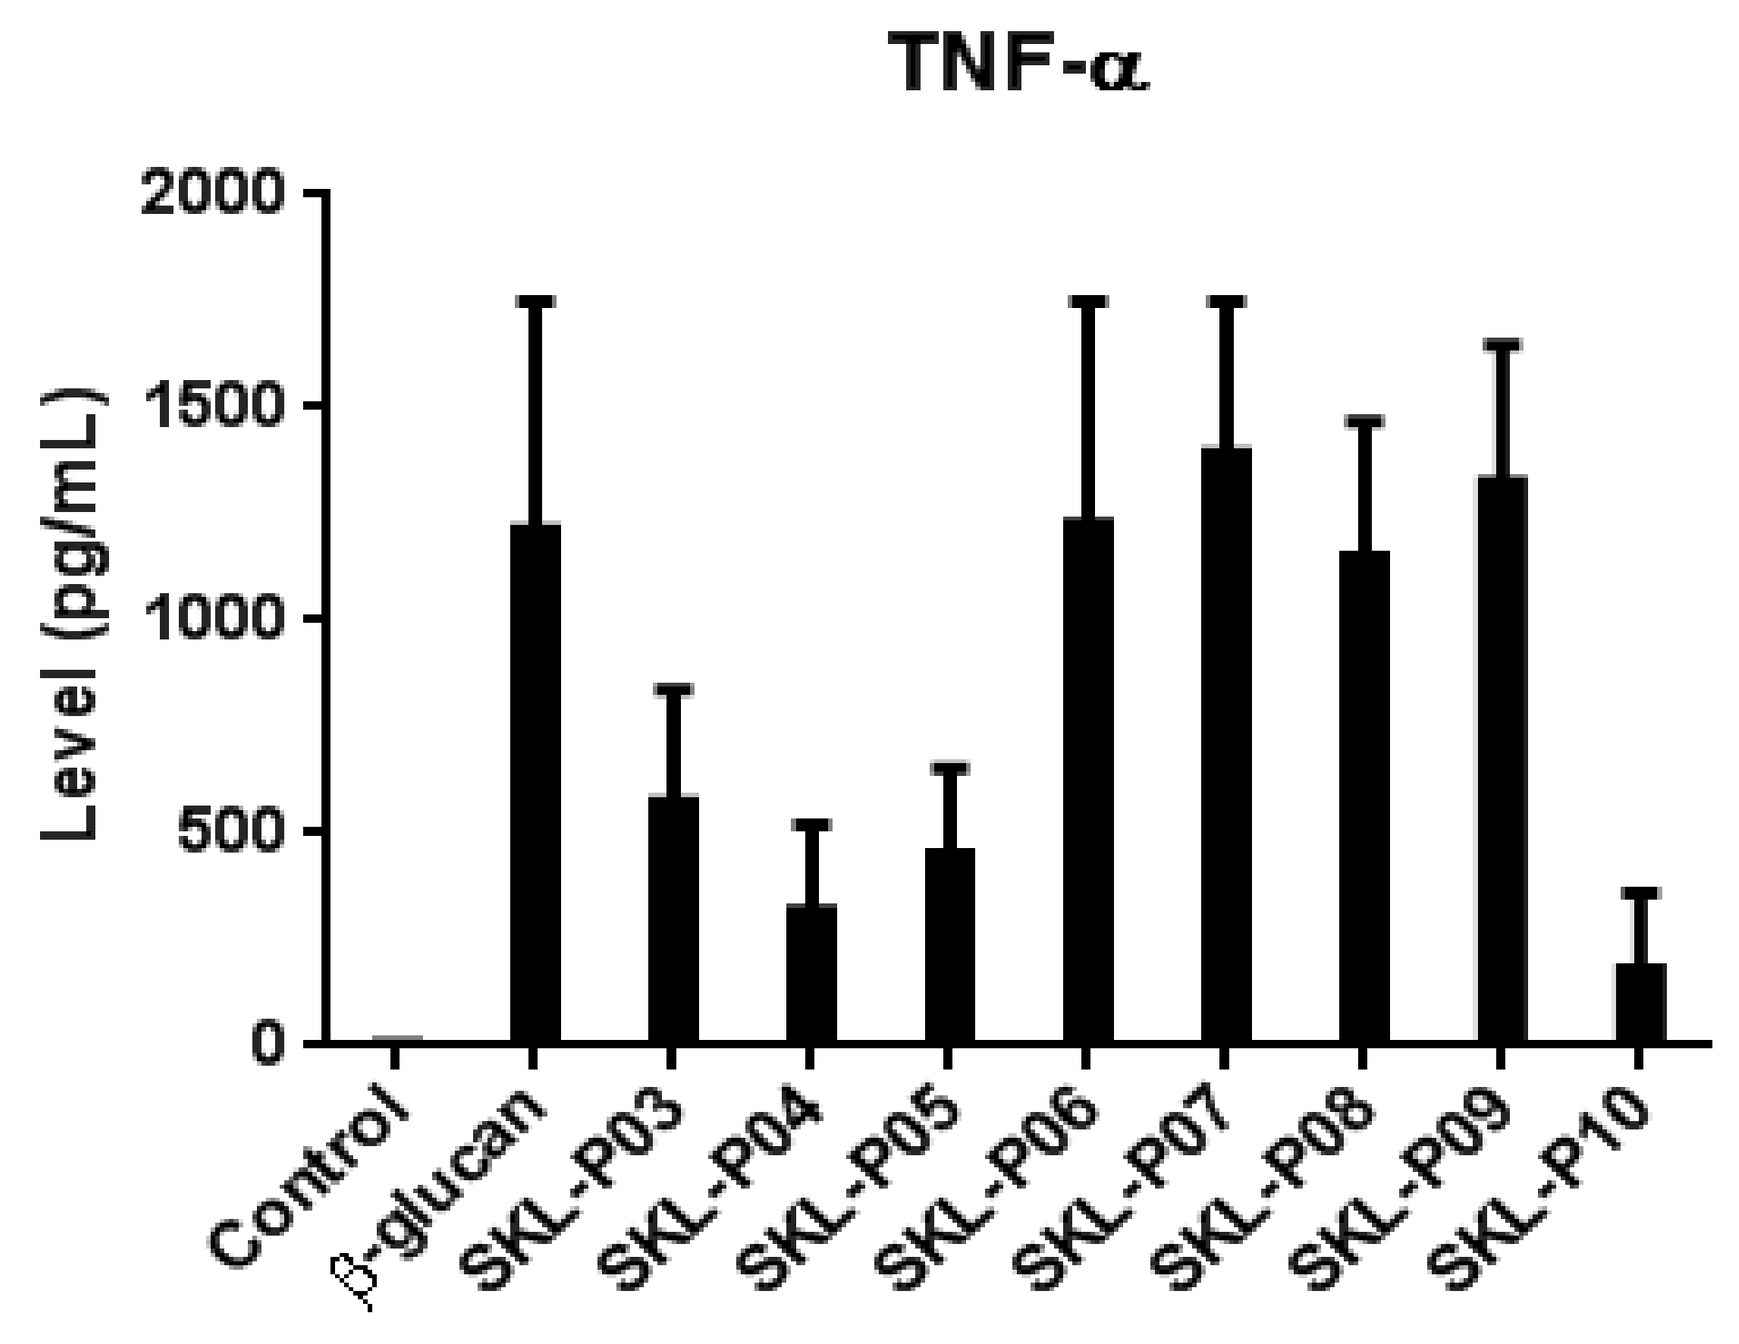
TNF-α
Level (pg/mL)
0 500 1000 1500 2000
Control
β-glucan
SKL-P03
SKL-P04
SKL-P05
SKL-P06
SKL-P07
SKL-P08
SKL-P09
SKL-P10

Figure 9.

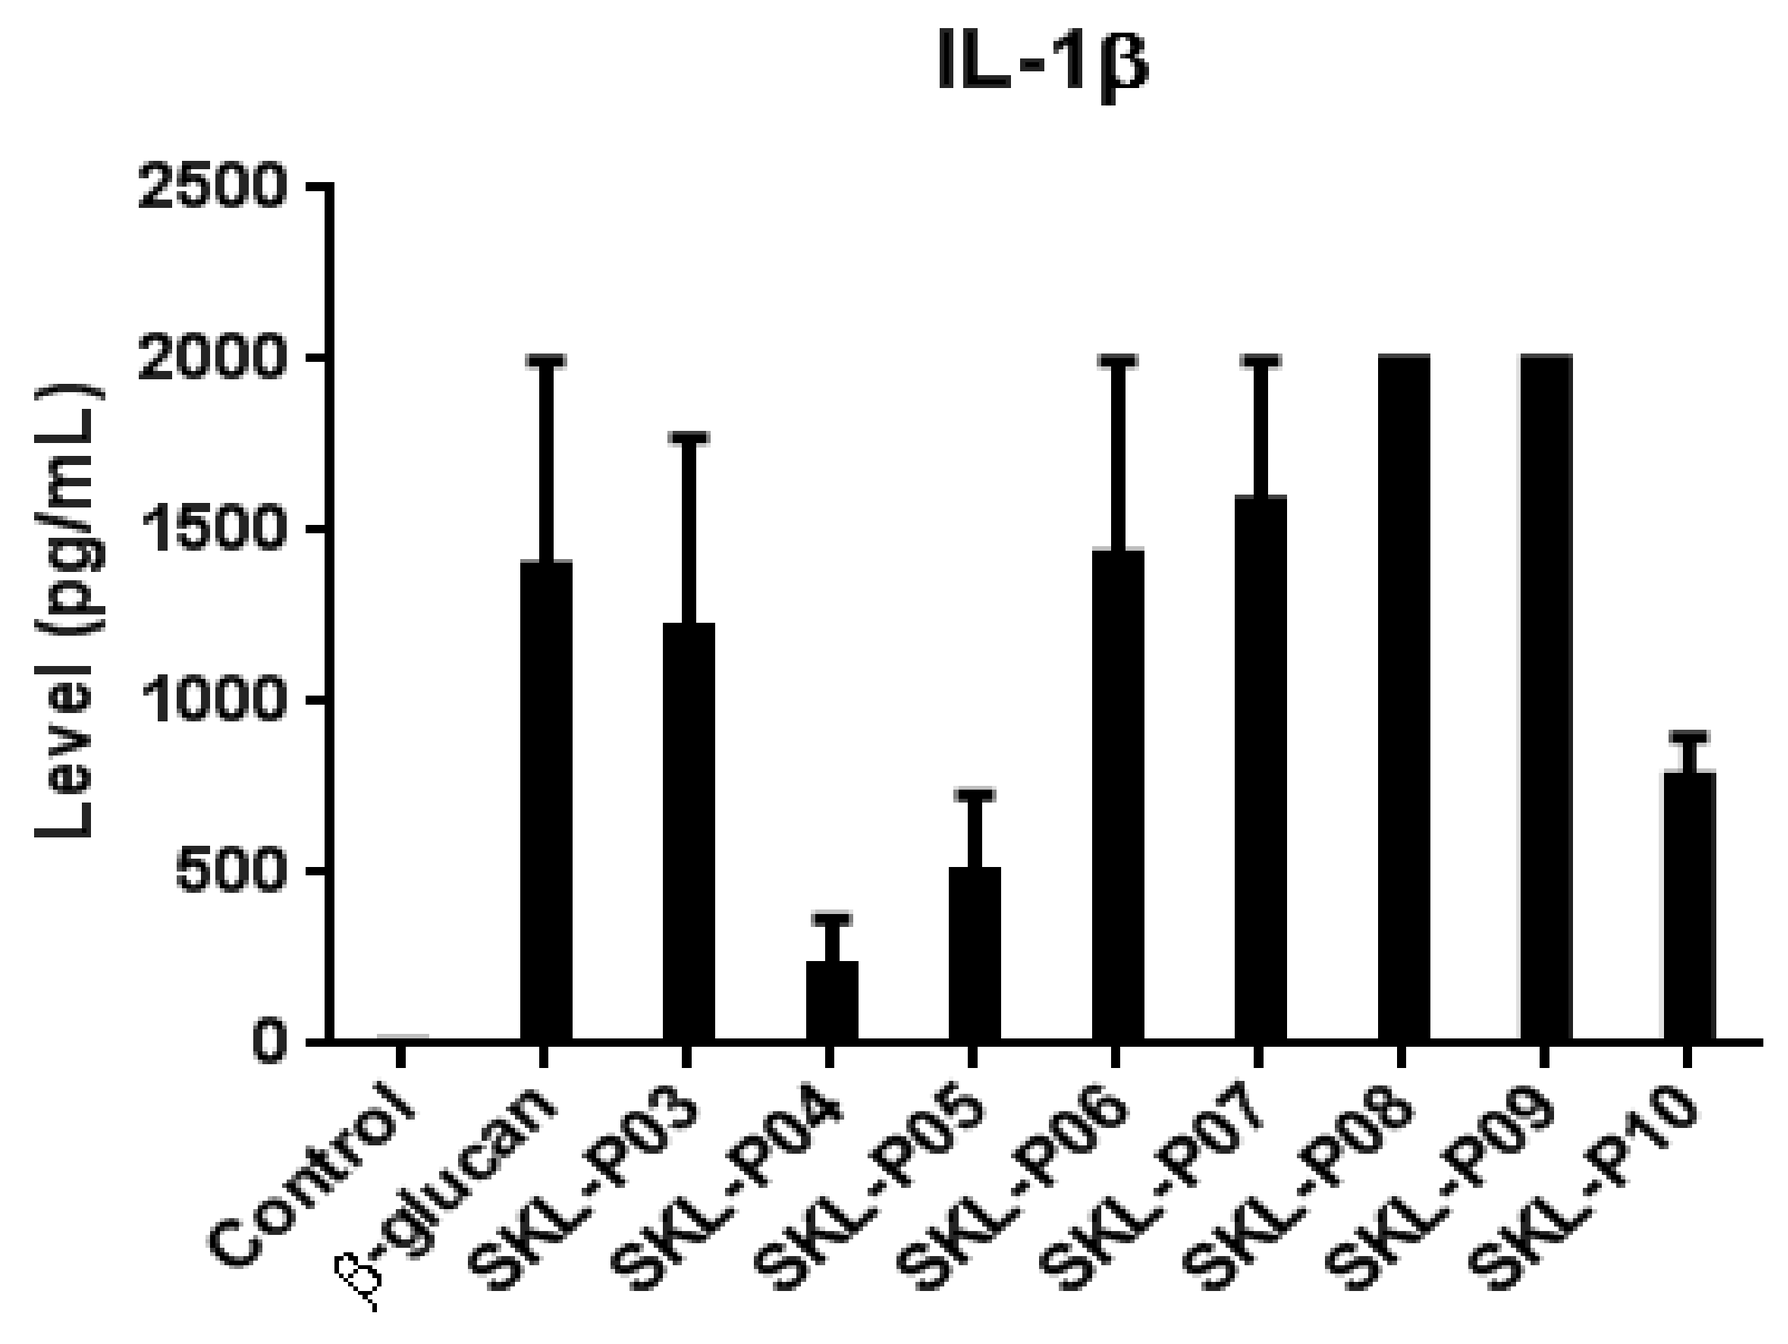
IL-1β
Level (pg/mL)
0 500 1000 1500 2000 2500
Control
β-glucan
SKL-P03
SKL-P04
SKL-P05
SKL-P06
SKL-P07
SKL-P08
SKL-P09
SKL-P10

Figure 10.

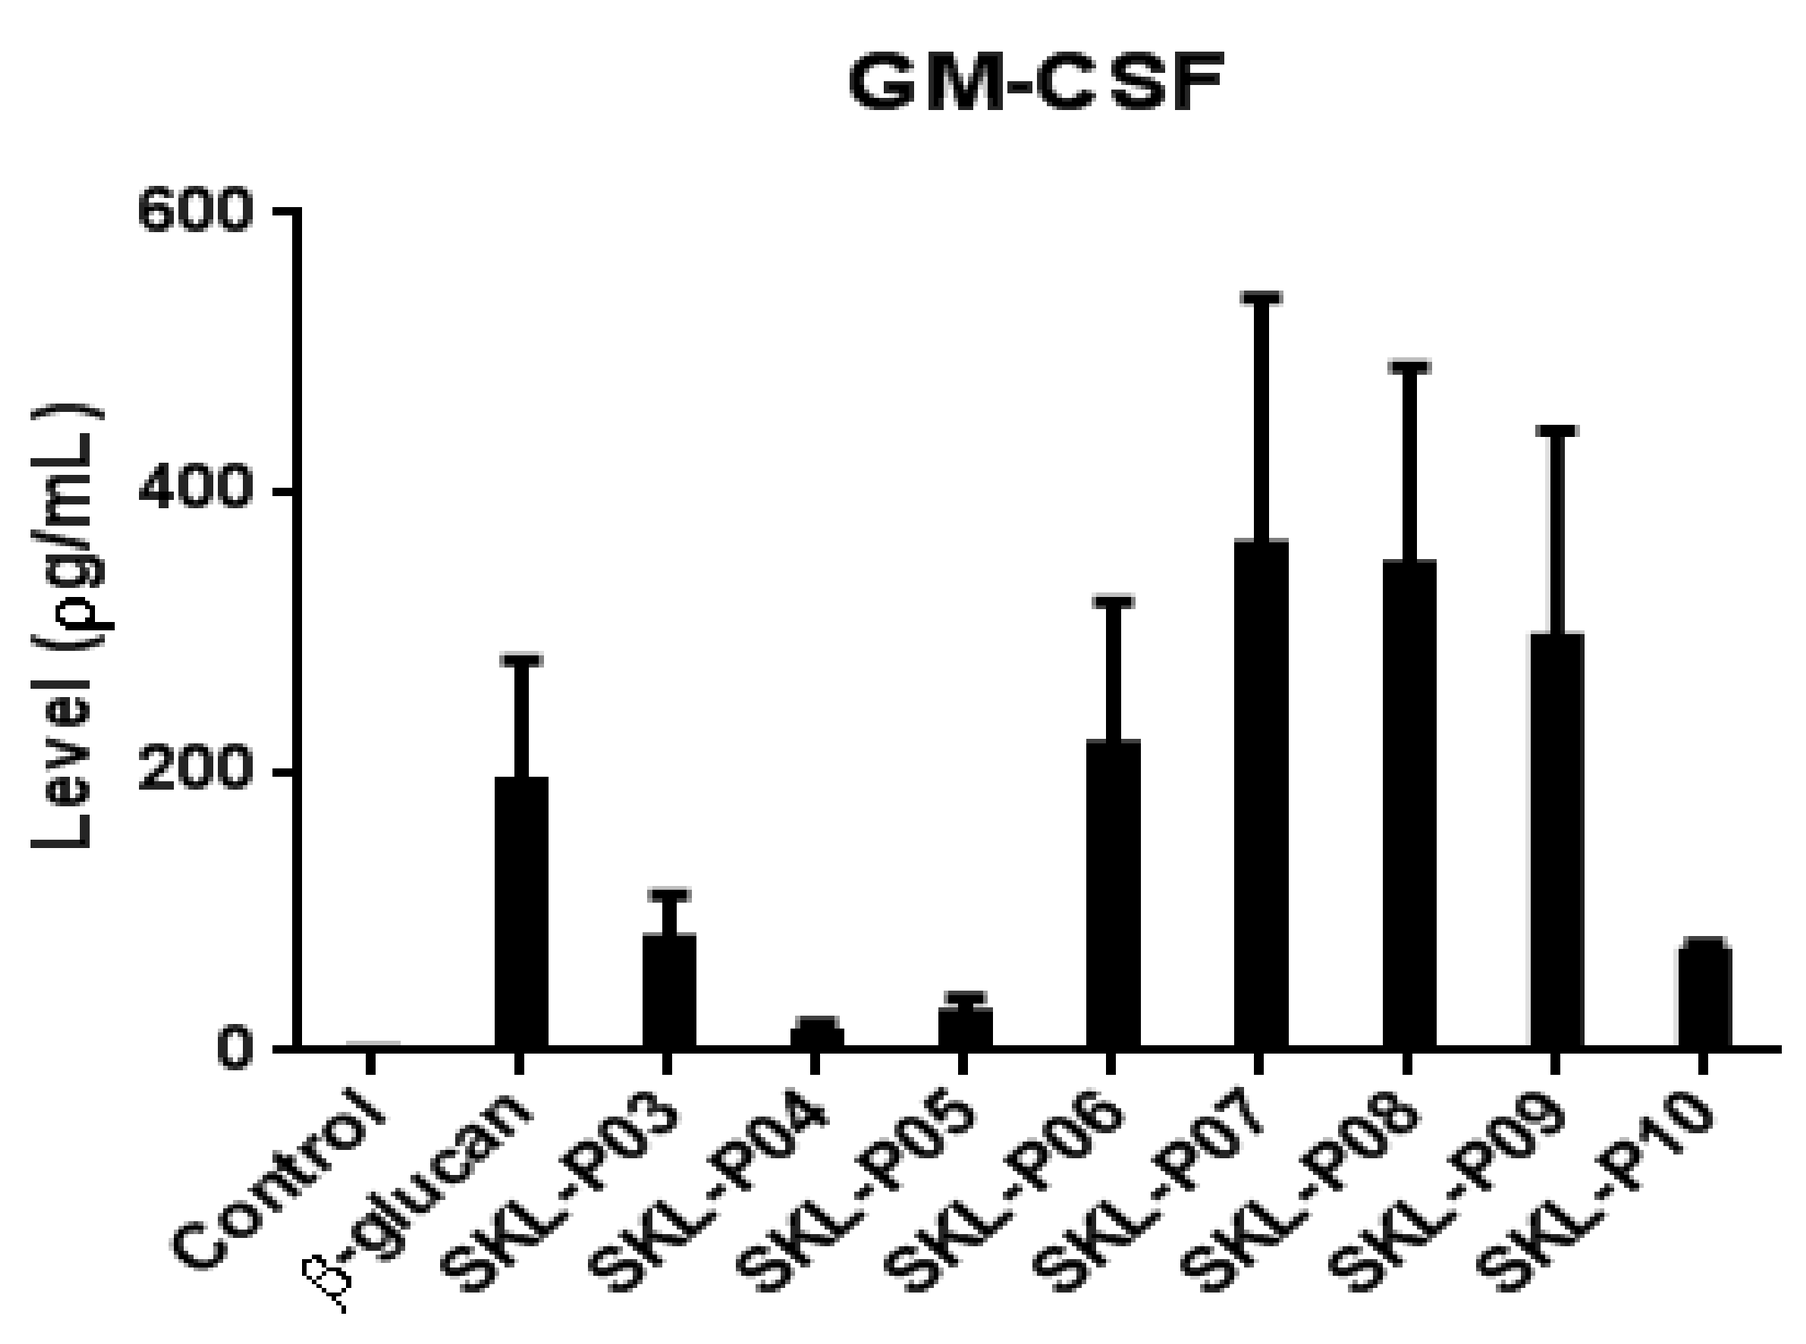
GM-CSF
Level (ρg/mL)
0 200 400 600
Control
β-glucan
SKL-P03
SKL-P04
SKL-P05
SKL-P06
SKL-P07
SKL-P08
SKL-P09
SKL-P10

(Abbreviation: IFN-γ: Interferon-gamma; IL-10: Interleukin-10; TNF-α: Tumor necrosis factor-alpha; IL-1β: Interleukin-1beta; GM-CSF: Granulocyte-macrophage colony-stimulating factor)

CONCLUSIONS

Our previous results showed that a specific yeast extract blend (PX3*) had potent immuno-stimulatory effects and was superior to yeast β-glucan alone in several cytokine parameters (IFN-γ, IL-10, IL-12) and lymphocyte proliferation. Additional experiments with different combinations of natural extracts (Panax ginseng/American ginseng, Princess Matsutake mushroom /Reishi mushroom, Astragalus root) showed interesting results. While some combination showed synergistic enhancement in activities, others showed reduced efficacy compared to β-glucan (10 µg/mL) alone. Based on our results, we were able to develop a dietary supplement that has the optimal combination of natural extracts (SKL-P09) to potentiate the immuno-stimulatory effects of β-glucan and enhance the efficacy.

REFERENCES

1. Samuelsen, A.B., J. Schrezenmeir, and S.H. Knutsen, *Effects of orally administered yeast-derived beta-glucans: a review.* Mol Nutr Food Res, 2014. **58**(1): p. 183-93.

2. Vannucci, L., et al., *Immunostimulatory properties and antitumor activities of glucans (Review).* Int J Oncol, 2013. **43**(2): p. 357-64.

3. Batbayar, S., D.H. Lee, and H.W. Kim, *Immunomodulation of Fungal beta-Glucan in Host Defense Signaling by Dectin-1.* Biomol Ther (Seoul), 2012. **20**(5): p. 433-45.

4. Hong, F., et al., *Mechanism by which orally administered beta-1,3-glucans enhance the tumoricidal activity of antitumor monoclonal antibodies in murine tumor models.* J Immunol, 2004. **173**(2): p. 797-806.

5. Yatawara, L., et al., *Aureobasidium-derived soluble branched (1,3-1,6) beta-glucan (Sophy beta-glucan) enhances natural killer activity in Leishmania amazonensis-infected mice.* Korean J Parasitol, 2009. **47**(4): p. 345-51.

6. Fuller, R., et al., *Influence of yeast-derived 1,3/1,6 glucopolysaccharide on circulating cytokines and chemokines with respect to upper respiratory tract infections.* Nutrition, 2012. **28**(6): p. 665-9.

7. McFarlin, B.K., et al., *Baker's yeast beta glucan supplementation increases salivary IgA and decreases cold/flu symptomatic days after intense exercise.* J Diet Suppl, 2013. **10**(3): p. 171-83.

8. Bergendiova, K., E. Tibenska, and J. Majtan, *Pleuran (beta-glucan from Pleurotus ostreatus) supplementation, cellular immune response and respiratory tract infections in athletes.* Eur J Appl Physiol, 2011. **111**(9): p. 2033-40.